Isolation and Identification of the Platelet-derived 8,9-11,12-Diepoxy-13-Hydroxy Eicosadienoic Acid (8,11-DiEp-13-HEDE)

Andrei Kornilov[1], Paul D. Kennedy[1], Maceler Aldrovandi[2], Andrew J.A. Watson[2], Christine Hinz[2], Bryan Harless[1], Joseph Colombo[1], Kirk M. Maxey[1], Victoria J. Tyrrell[2], Matthew Simon[1], Varinder K. Aggarwal[3], William E. Boeglin[4], Alan R. Brash[4], Robert C. Murphy[5], Valerie B. O'Donnell[2]

[1]Cayman Chemical, Ann Arbor, MI, United States; [2]Cardiff University, United Kingdom; [3]University of Bristol, United Kingdom; [4]Vanderbilt University Medical School, United States; [5]University of Colorado Health Sciences Center, United States

KEY FINDINGS

A novel eicosanoid from arachidonic acid oxidation has been isolated and characterized as 8,11-DiEp-13-HEDE.

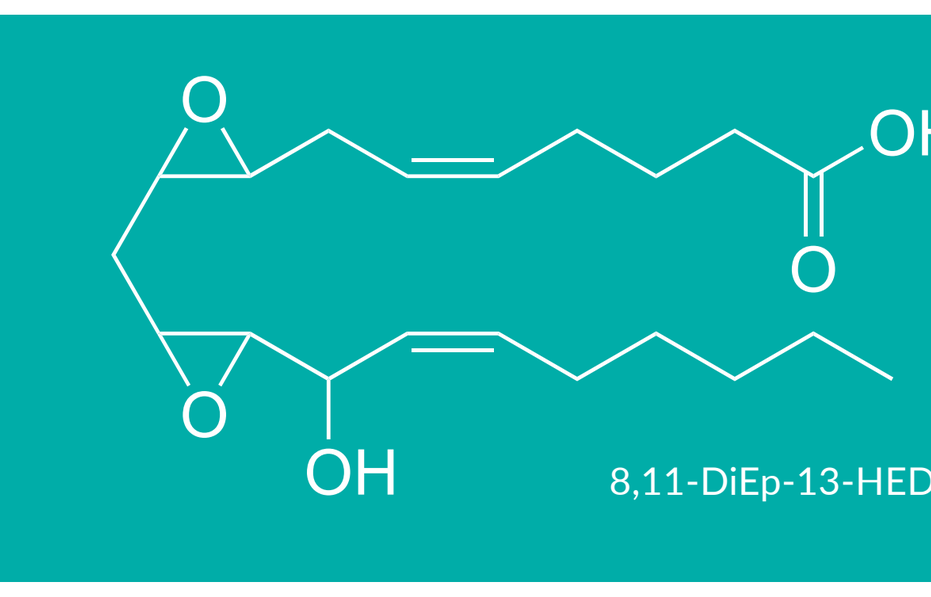
O
OH
O
O
OH
8,11-DiEp-13-HEDE

Aims of the Study

Recent studies into platelet-derived oxidation products of arachidonic acid have led to the discovery of a novel eicosanoid.[1] In this study, we set out to isolate this novel eicosanoid to confirm its structure and ultimately evaluate its biological activity and function.

Background

Oxygenase enzymes in platelets have been previously shown to produce a variety of arachidonic acid-derived oxylipins including endoperoxides and hydroxy-diepoxides.[2,3] O'Donnell *et al.* identified a new eicosanoid in thrombin-activated human platelets.[1] In collaboration with the O'Donnell lab, scientists at Cayman Chemical attempted to isolate this eicosanoid using non-enzymatic oxidation. The original publication suggested using 11-HpETE, but we later determined that arachidonic acid could be used with the same outcome (see below).

Synthetic Route

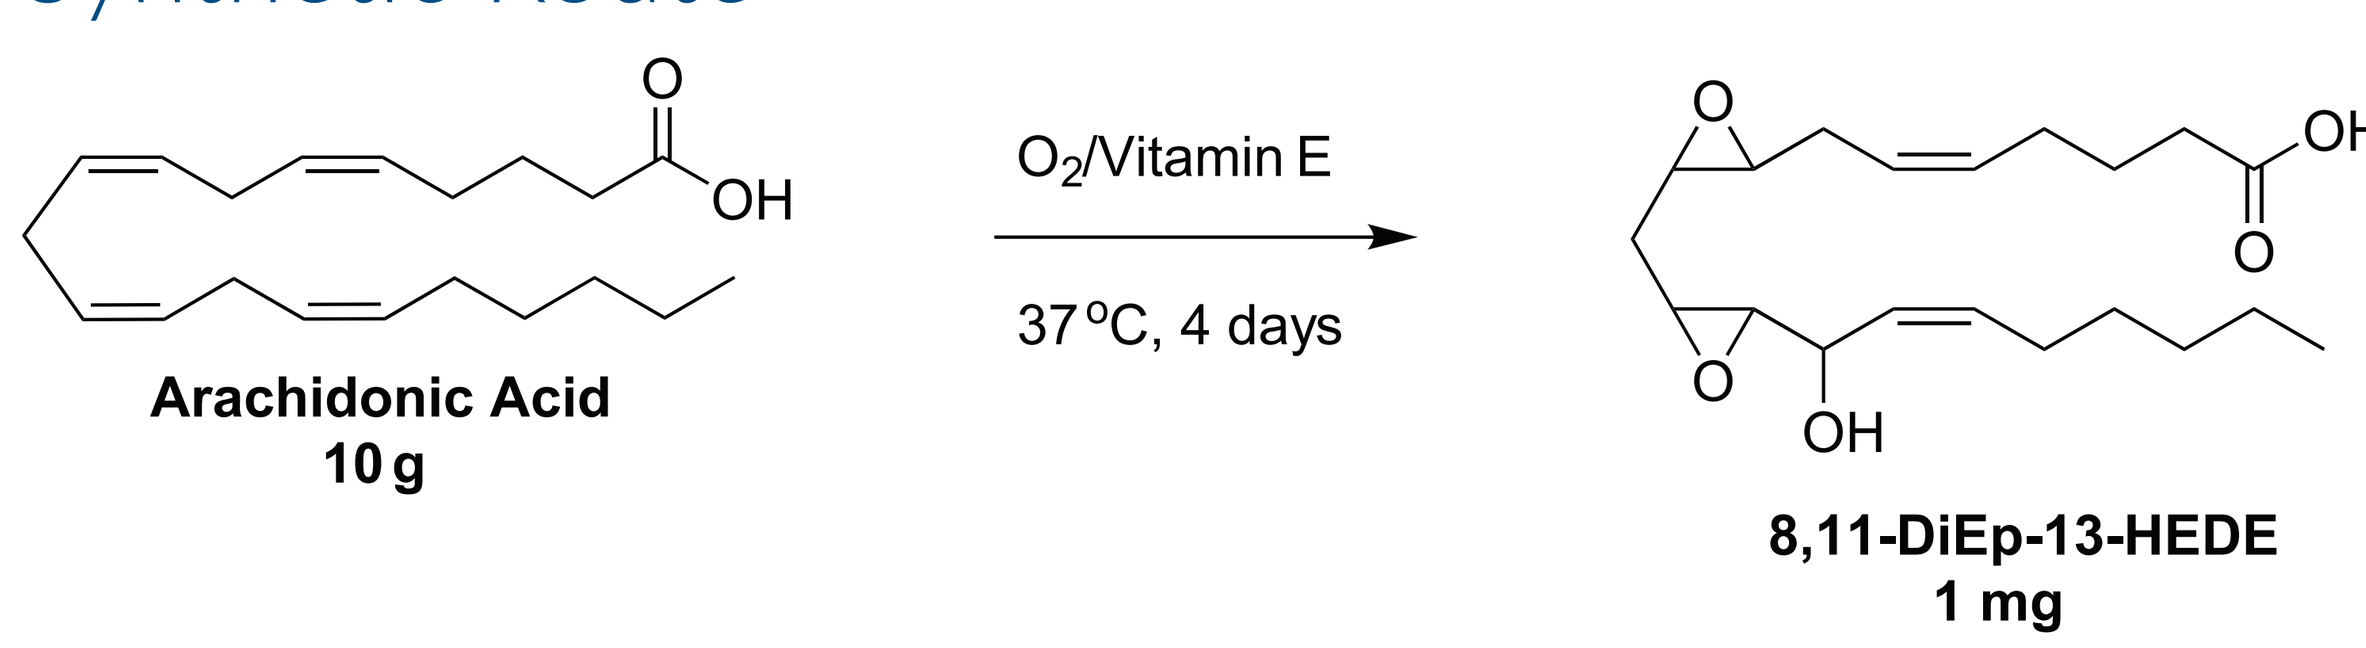
O
OH
Arachidonic Acid
10 g
O2/Vitamin E
37°C, 4 days
O
OH
O
O
OH
8,11-DiEp-13-HEDE
1 mg

Scheme 1. Non-enzymatic oxidation of arachidonic acid produced a small oxidized eicosanoid possessing the same mass spectrum and retention time as the original platelet-derived DXA_3.

Analytical LC-MS/MS Method Details

All mass spectrometric data was acquired on UltiMate 3000 UHPLC (Thermo Scientific™) coupled to a Q Exactive™ mass spectrometer (Thermo Scientific™) with the following conditions: Electrospray ionization was used in negative mode, with a sheath gas flow rate of 60 units, and capillary temperature at 320°C. Data-dependent MS/MS of *m/z* 351 was carried out with a resolving power of 35,000.

LC-MS/MS Method 1

A: 0.1% Formic acid in water
B: 0.1% Formic acid in acetonitrile
Gradient:
- 0-45 min 43.7% B
- 55 min 100% B
- 65 min 100% B
- 65.1 min 43.7% B
- 75 min 43.7% B

Luna Phenyl-Hexyl 5 µm
250 x 4.6 mm 1 ml/min

LC-MS/MS Method 2

A: 0.1% Formic acid in water
B: 0.1% Formic acid in acetonitrile
Gradient:
- 0-1 min 15-28% B
- 1-4 min 28-42% B
- 4-6 min 42-60% B
- 6-16 min 60-80% B
- 16-19 min 80-95% B
- 19-22 min 95% B
- 22.1-25.1 min 15% B

Phenomenex Kinetex C18 2.6 µm
100 x 2.1 mm 400 µl/min

Flash Chromatography Method

Silica gel hexane:ethyl acetate:acetic acid, 90:10:0.5–50:50:0.5

Semi-preparative HPLC Method

95:5:0.1 Heptane:IPA:acetic acid
Luna Silica 5 µm
250 x 10 mm 5 ml/min monitored at 206 nm

Analytical HPLC Method

97:2.5:0.5:0.1 Heptane:IPA:MeOH:acetic acid
Luna Silica 5 µm
250 x 4.6 mm 0.5 ml/min monitored at 206 nm

Identification and Purification

The target compound had an *m/z* of 351.2 and a characteristic fragment of 165.1 *m/z*. This peak was targeted for purification and identification.

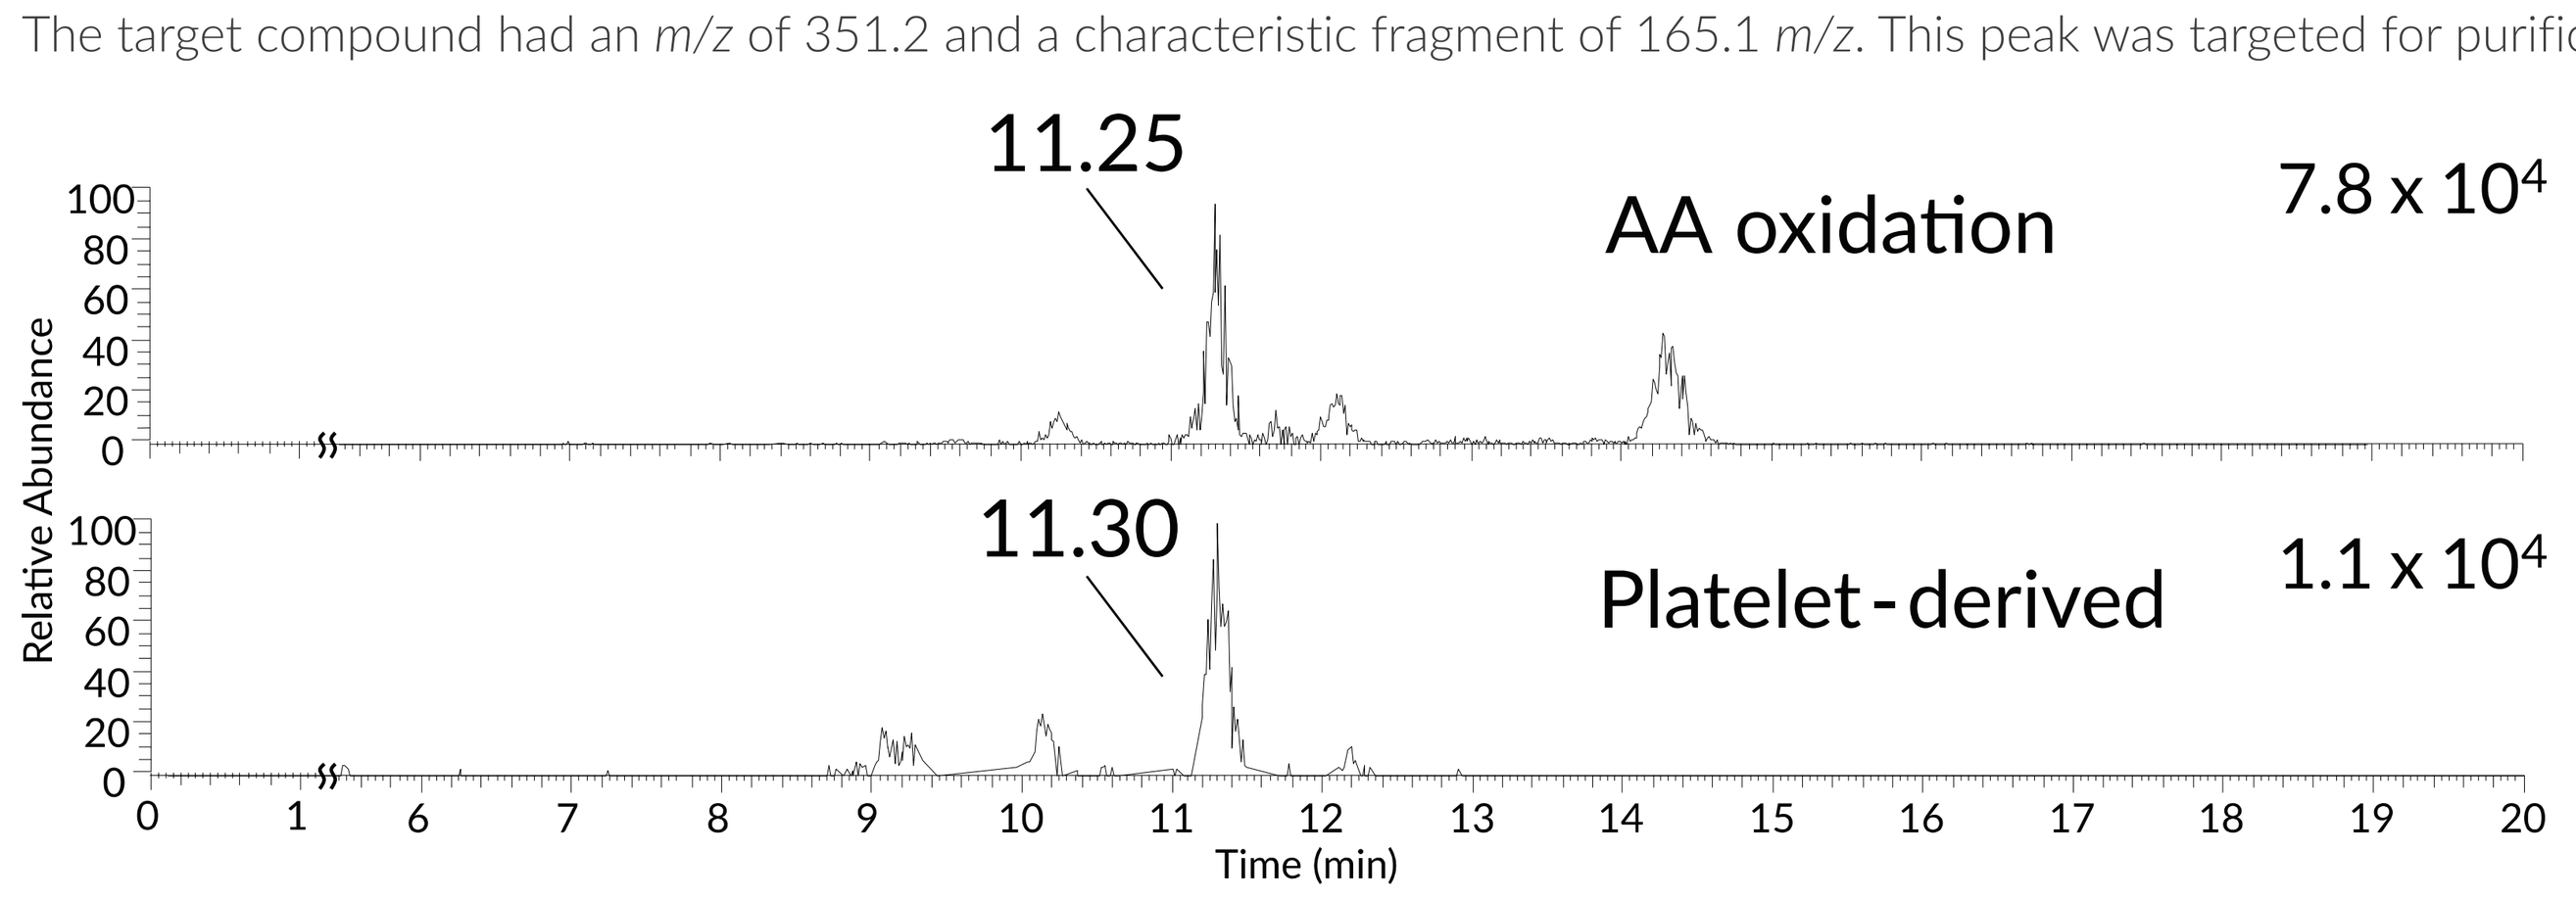
11.25
AA oxidation
7.8 x 10^4
11.30
Platelet-derived
1.1 x 10^4
Relative Abundance
Time (min)

Figure 1. Extracted ion chromatogram of the 165.1 product ion generated from MS/MS of the precursor 351.2 ion using LC-MS/MS Method 1.

Learn more about what our Analytical Services team can do for you at **www.caymanchem.com/analyticalchem**

References
1. Hinz, C., Aldrovandi, M., Uhlson, C., *et al.* Human platelets utilize cycloxygenase-1 to generate dioxolane A_3, a neutrophil-activating eicosanoid. *J. Biol. Chem.* **291(26)**, 13448-13464 (2016).
2. Teder, T., Boeglin, W.E., and Brash, A.R. Lipoxygenase-catalyzed transformation of epoxy fatty acids to hydroxy-endoperoxides: A potential P450 and lipoxygenase interaction. *J. Lipid Res.* **55(12)**, 2587-2596 (2014).
3. Schneider, C., Boeglin, W.E., and Brash, A.R. Identification of two cyclooxygenase active site residues, leucine 384 and glycine 526, that control carbon ring cyclization in prostaglandin biosynthesis. *J. Biol. Chem.* **279(6)**, 4404-4414 (2004).

HPLC Purification Workflow and Yield

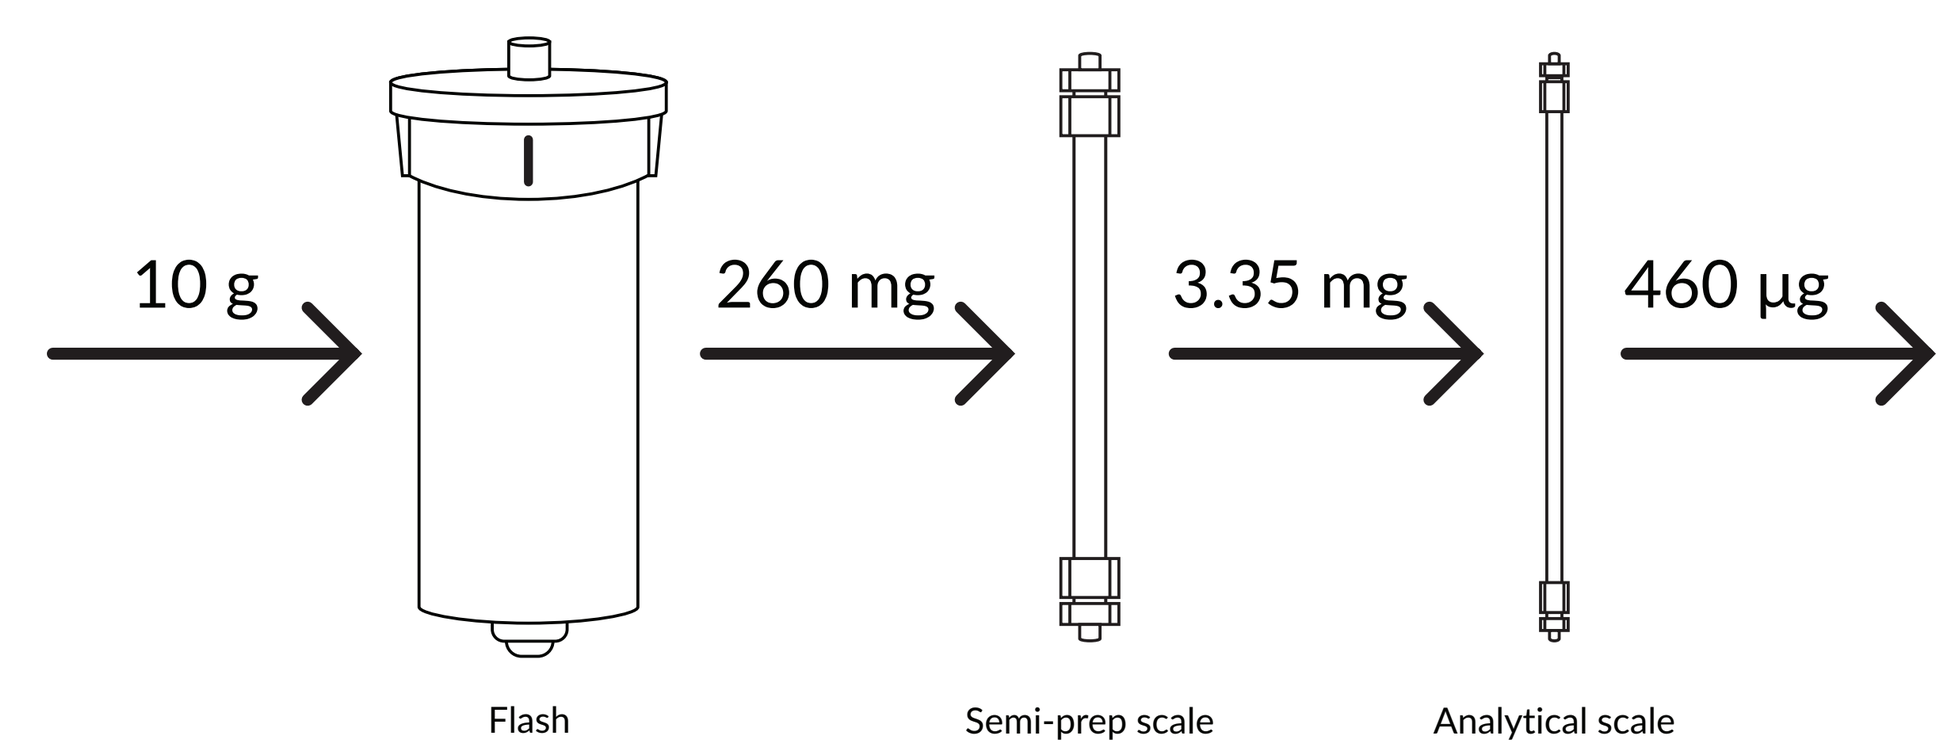
10 g
260 mg
3.35 mg
460 µg
Flash
Semi-prep scale
Analytical scale

Structural Elucidation

MS/MS Match to Platelet Lipid

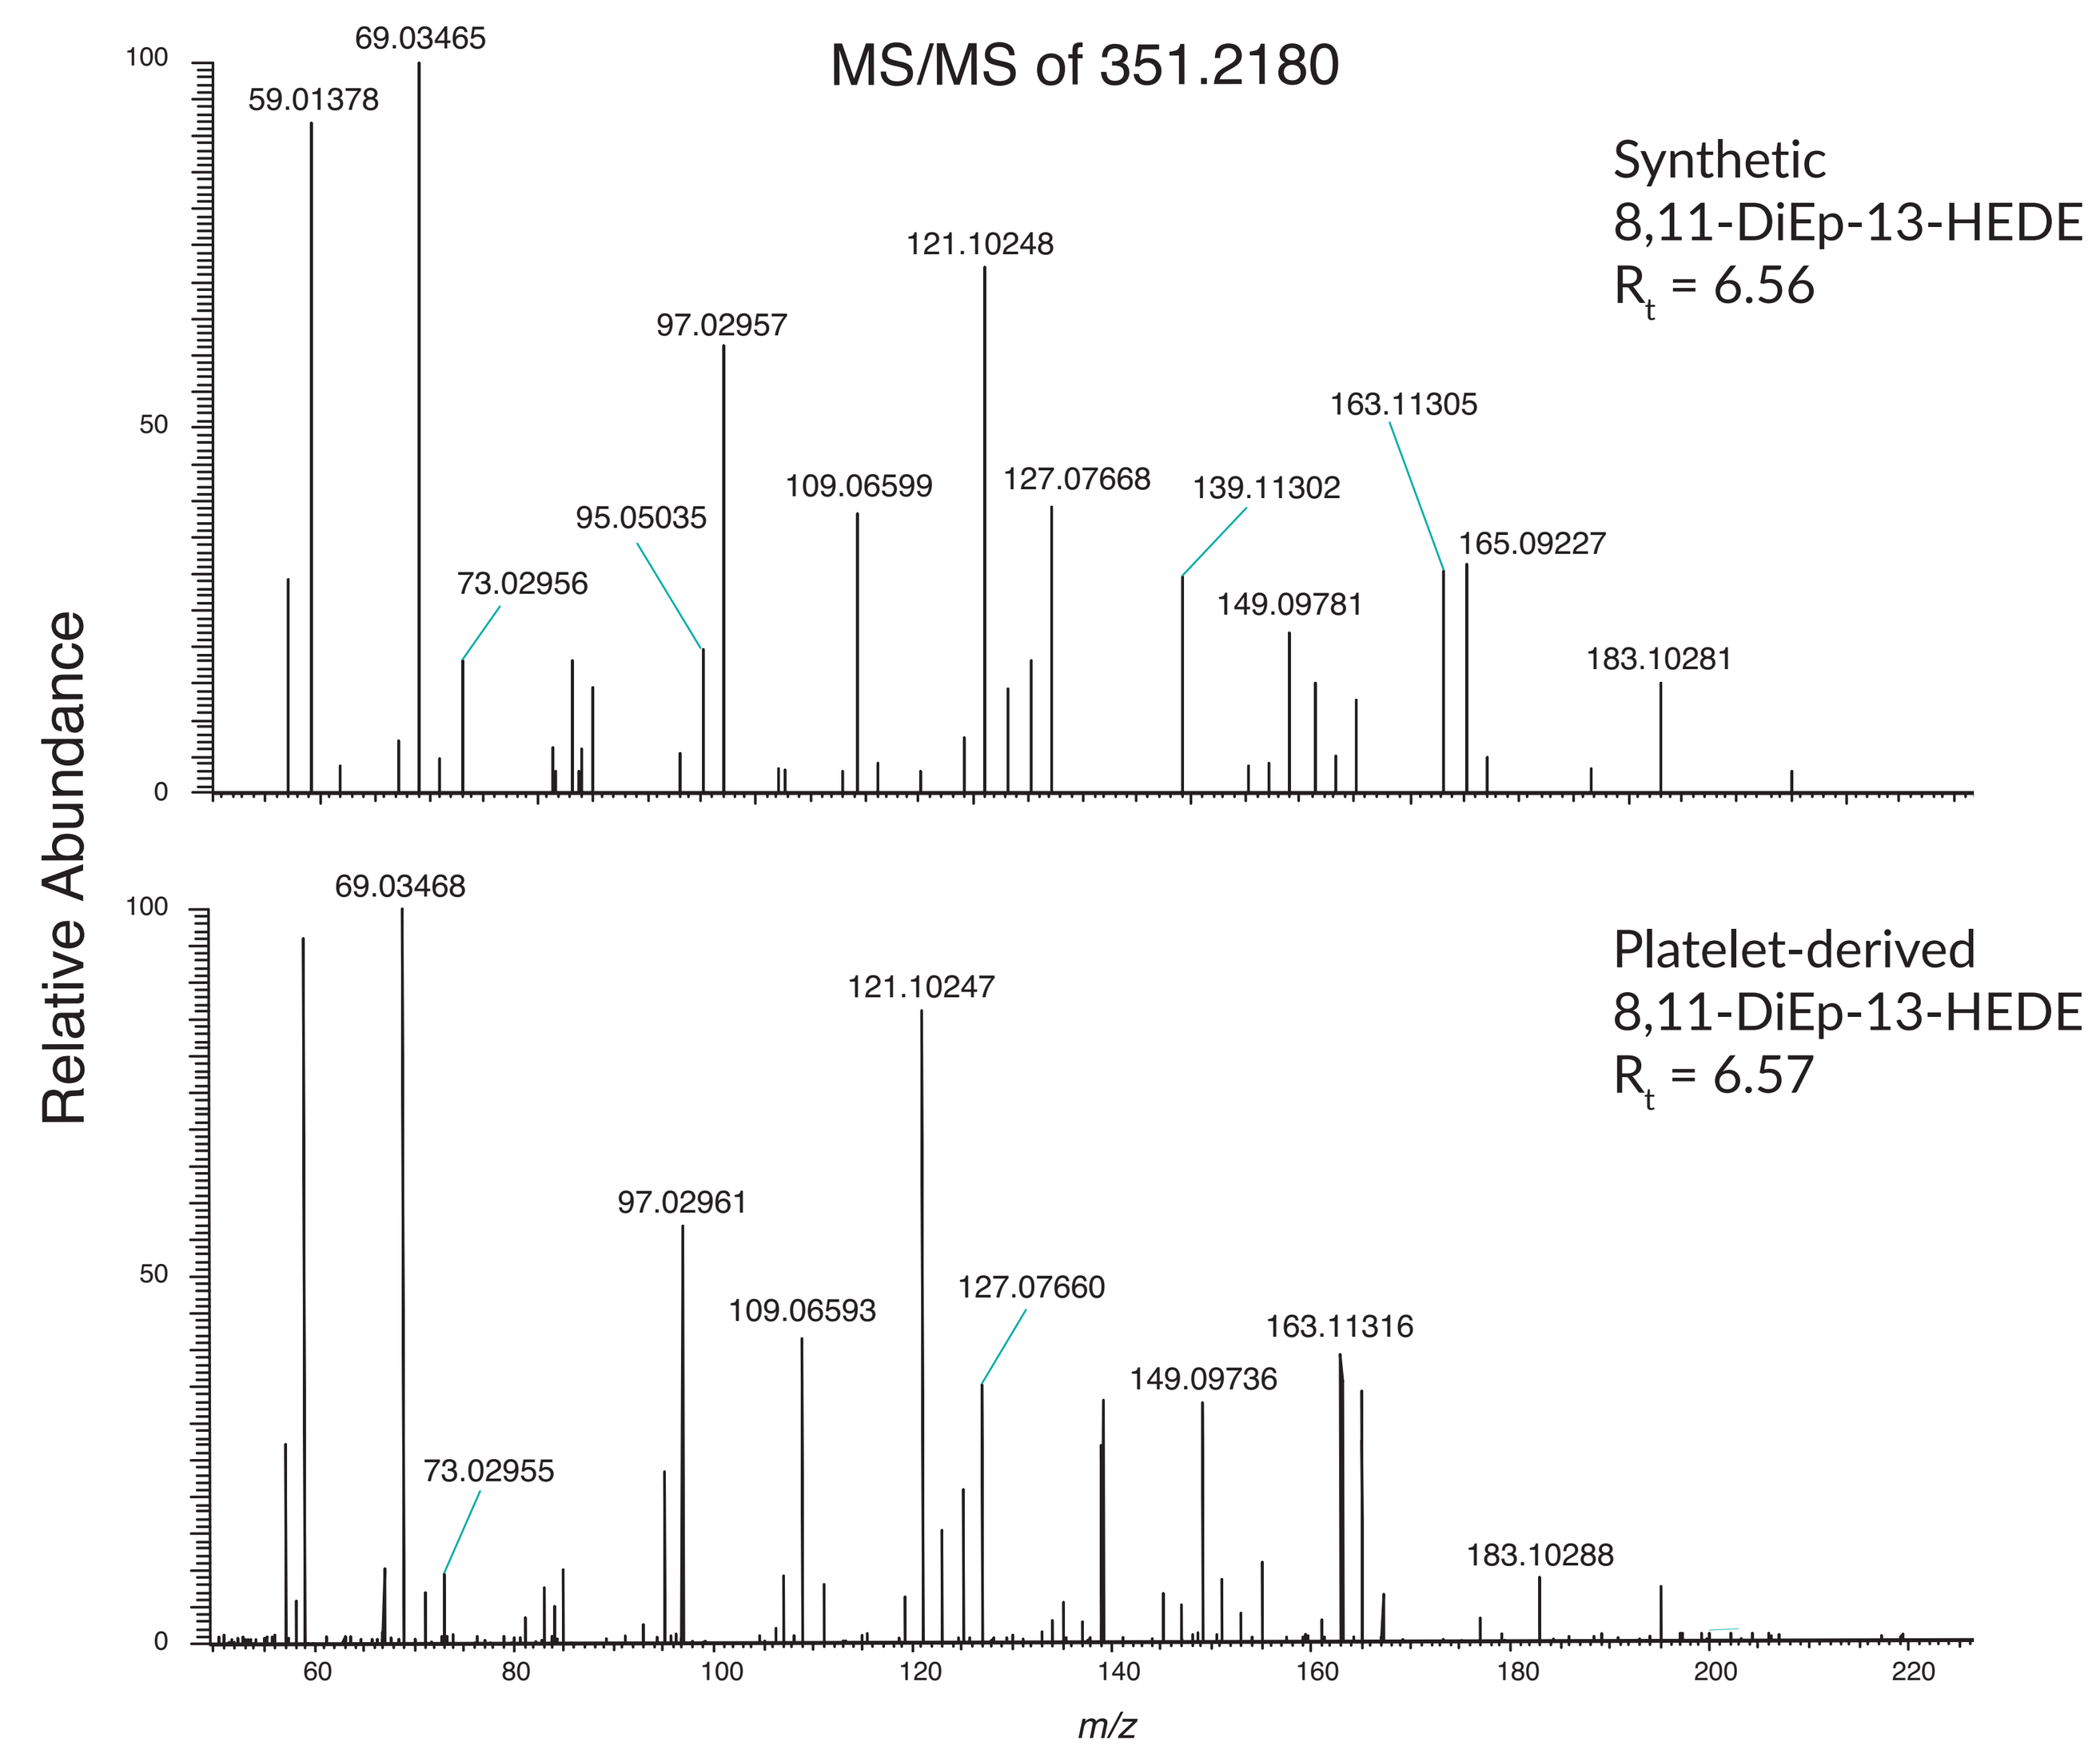
MS/MS of 351.2180
Synthetic
8,11-DiEp-13-HEDE
Rt = 6.56
69.03465
59.01378
121.10248
97.02957
163.11305
109.06599
127.07668
139.11302
95.05035
165.09227
73.02956
149.09781
183.10281
Platelet-derived
8,11-DiEp-13-HEDE
Rt = 6.57
69.03468
121.10247
97.02961
109.06593
127.07660
163.11316
149.09736
73.02955
183.10288
Relative Abundance
m/z

Product ion spectra of 351.2180 comparison of purified synthetic product (top) and natural extracted product (bottom) using **LC-MS/MS Method 2**. The presence of the target lipid or related isomer in the arachidonic oxidation reaction was confirmed *via* LC-MS/MS retention time, high-resolution precursor ion mass, and product ion spectrum.

Figure 2. Comparison of platelet DXA_3 to arachidonic acid autoxidation. Closely matching retention times (top) and product ion spectra (bottom) suggested that the synthetic lipid was either DXA_3 or a closely related isomer.

¹H NMR

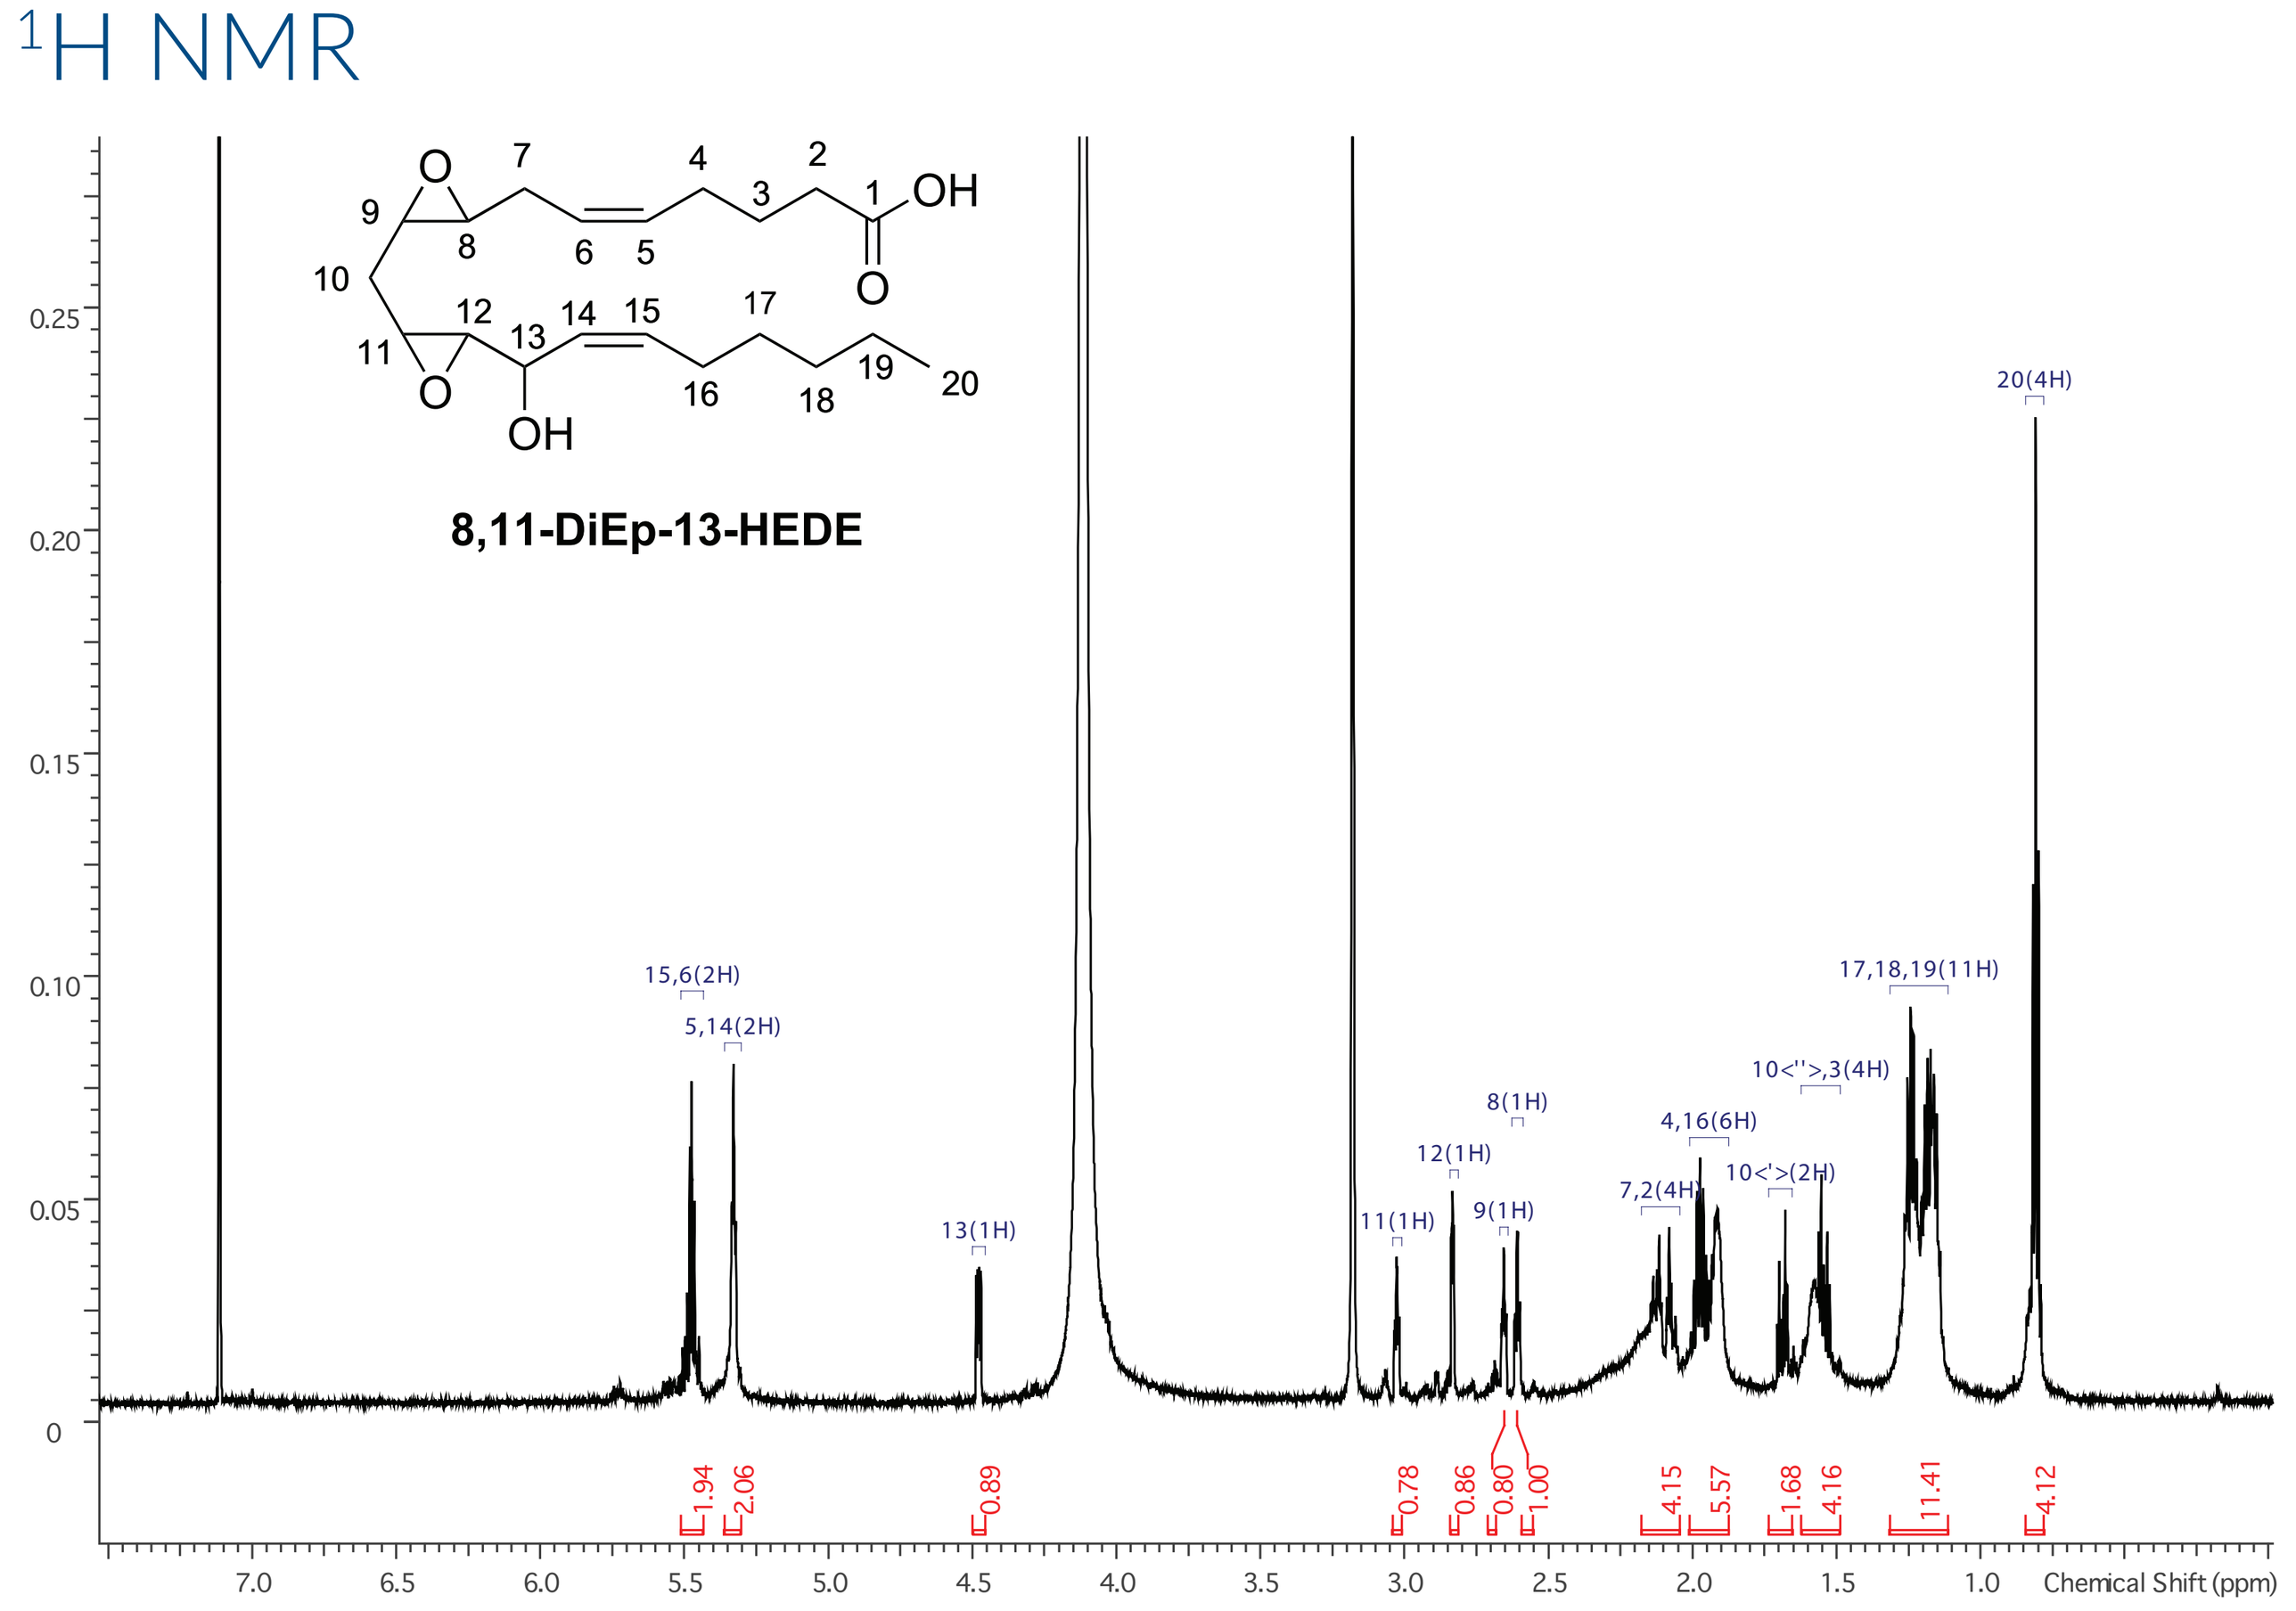
8,11-DiEp-13-HEDE
15,6(2H)
5,14(2H)
13(1H)
11(1H)
12(1H)
8(1H)
9(1H)
7,2(4H)
4,16(6H)
10<'>(2H)
10<''>,3(4H)
17,18,19(11H)
20(4H)
Chemical Shift (ppm)

The ¹H NMR experiment provides the following structural information:
- 4 double bond protons
- 4 epoxide protons
- 1 CH-OH proton
- 21 aliphatic protons including a CH_2 triplet adjacent to a carboxylic acid

Figure 3. ¹H NMR of purified target lipid. ¹H NMR (benzene-d_6/methanol-d_4 90:10, Varian 700 MHz). The 90:10 mix was used to resolve the epoxide proton signals without losing solubility.

¹H-¹H COSY

The ¹H-¹H COSY experiment provides data which supports the following structural details:
- Epoxy rings are separated by H10'-H10" CH_2
- The 13-C**H**-OH is adjacent to one double bond and one epoxy ring
- A chain of three aliphatic methylene CH_2s is between the COOH and first double bond
- There is a methylene between the first double bond and first epoxy ring

Based on this information, the structure proposed in **Figure 3** was determined to be the best fit for the data.

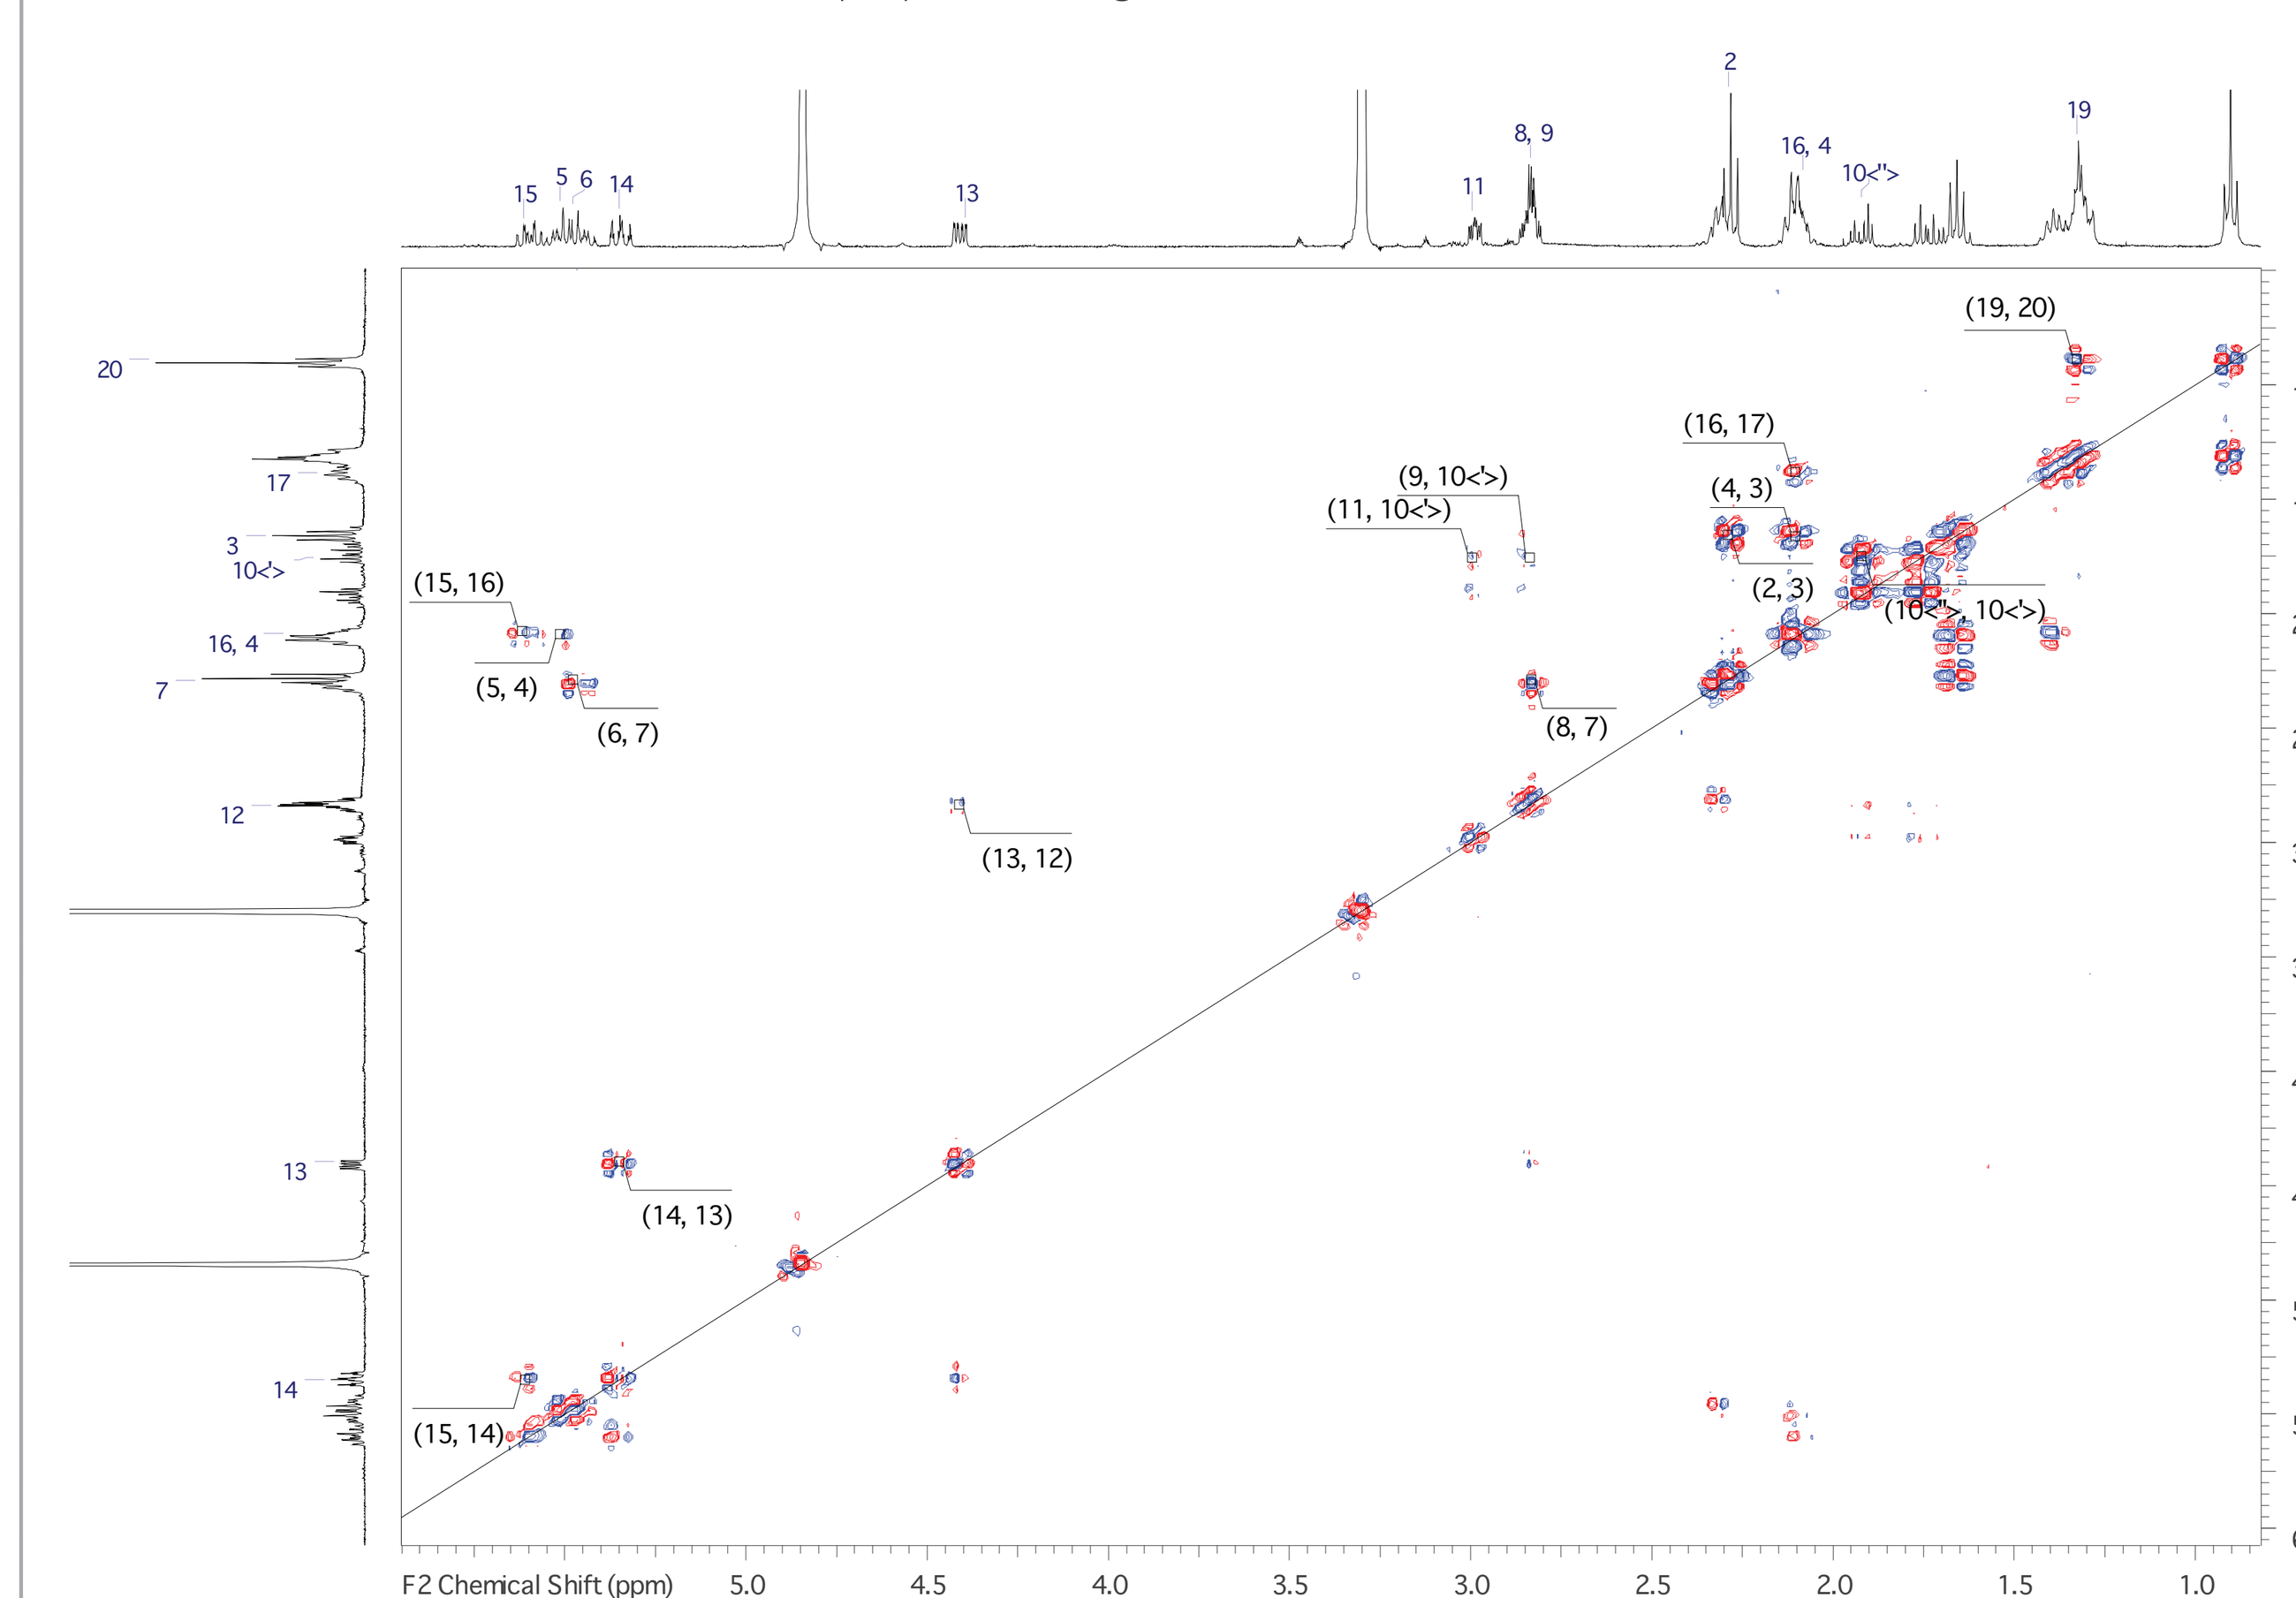
(19, 20)
(16, 17)
(9, 10<'>)
(11, 10<'>)
(4, 3)
(15, 16)
(2, 3)
(10<''>, 10<'>)
(5, 4)
(6, 7)
(8, 7)
(13, 12)
(14, 13)
(15, 14)
F2 Chemical Shift (ppm)

Figure 4. ¹H-¹H COSY spectrum of purified lipid.

NMR Assignments

F2 Axis (ppm)	F1 Axis (ppm)	Correlation
1.33	0.89	H20-19
1.38	1.33	H18-17
1.65	2.29	H3-2
1.66	2.08	H4-3
1.92	1.76	H10'-10"
2.1	1.38	H17-16
2.83	2.29	H8-7
2.84	1.93	H10'-9
2.84	1.76	H10"-9
3.00	1.90	H11-10'
3.00	1.76	H11-10"
4.41	2.84	H13-12
5.35	4.39	H14-13
5.48	2.29	H7-6
5.51	2.09	H5-4
5.61	5.35	H15-14
5.62	2.08	H16-15

Configuration of Epoxides

A.

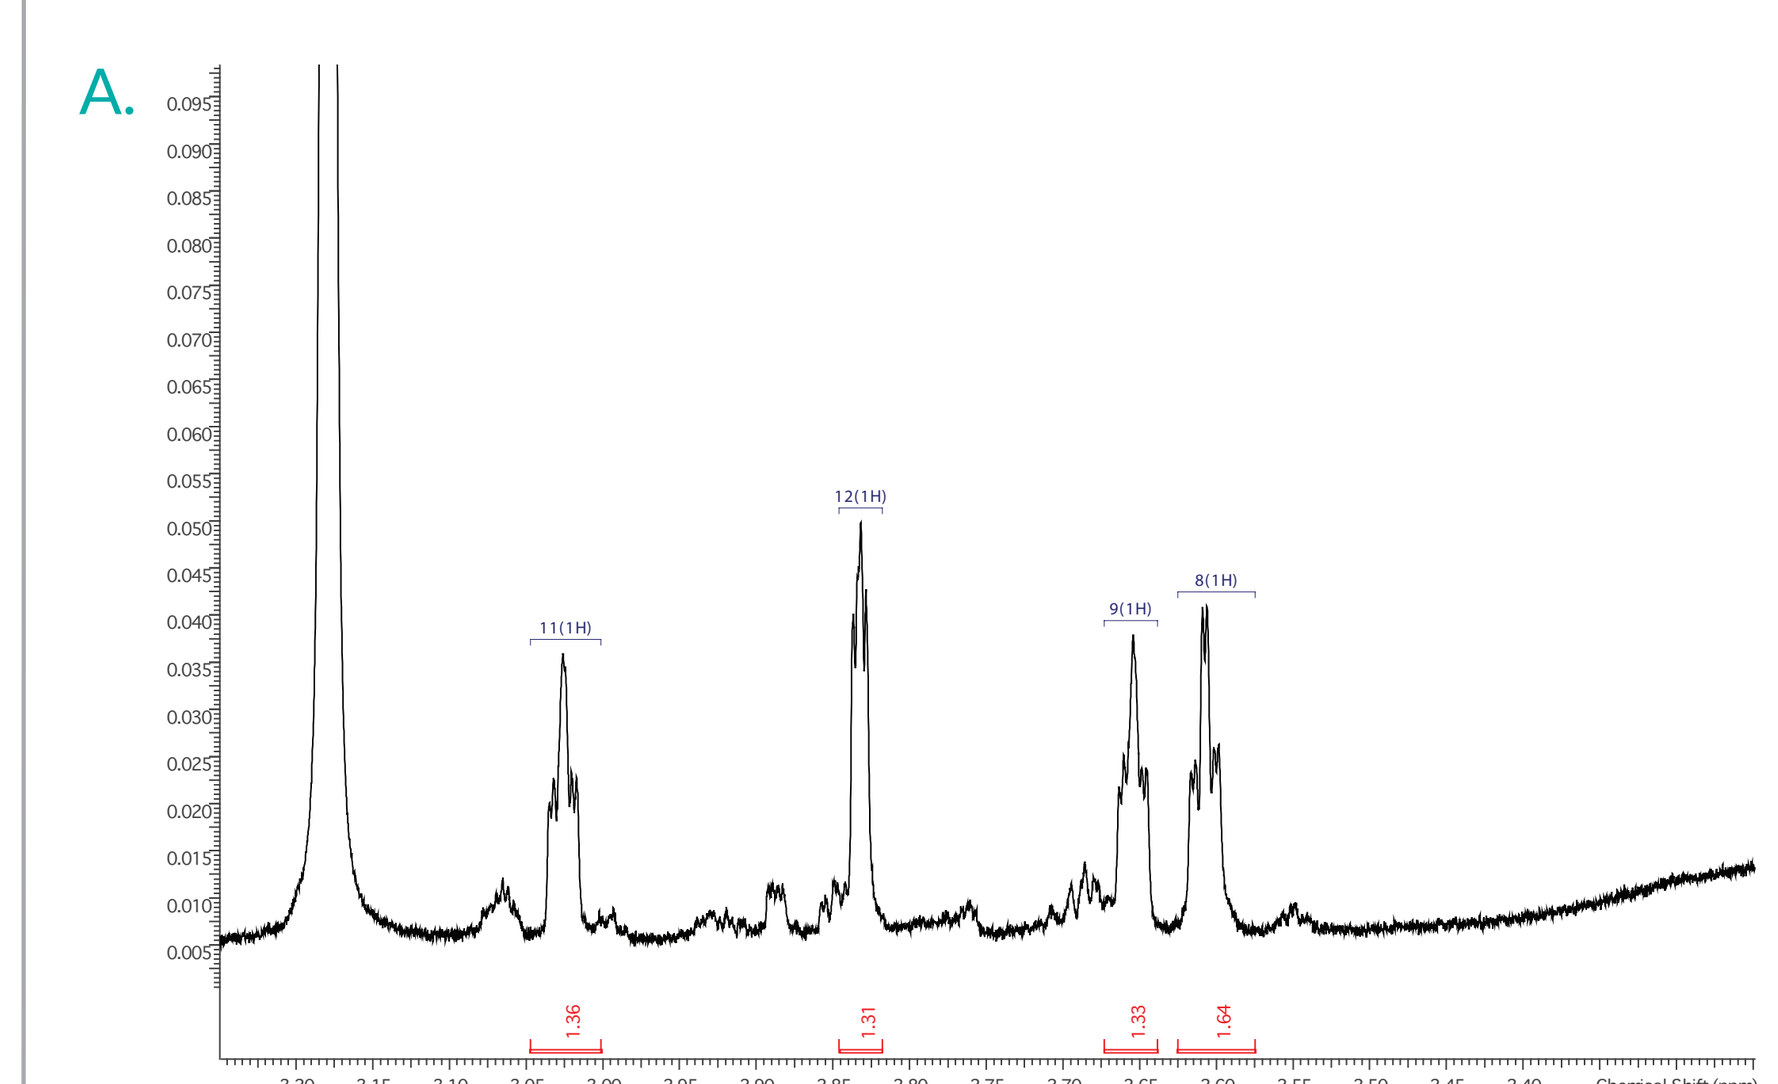
11(1H)
12(1H)
9(1H)
8(1H)
Chemical Shift (ppm)

Chemical Shift (ppm)	Multiplicity	Proton Carbon Number	Coupling Constant (Hz)
4.478	dd	H13	J_{13-14} = 7.75, J_{12-13} = 4.0
3.026	dt	H11	J_{11-12} = 2.20, J_{10-11} = 5.1
2.833	dd	H12	J_{11-12} = 2.13, J_{12-13} = 3.83
2.655	dt	H9	J_{8-9} = 2.25, J_{9-10} = 5.2
2.608	dt	H8	J_{8-9} = 2.21, J_{7-8} = 5.36

B.

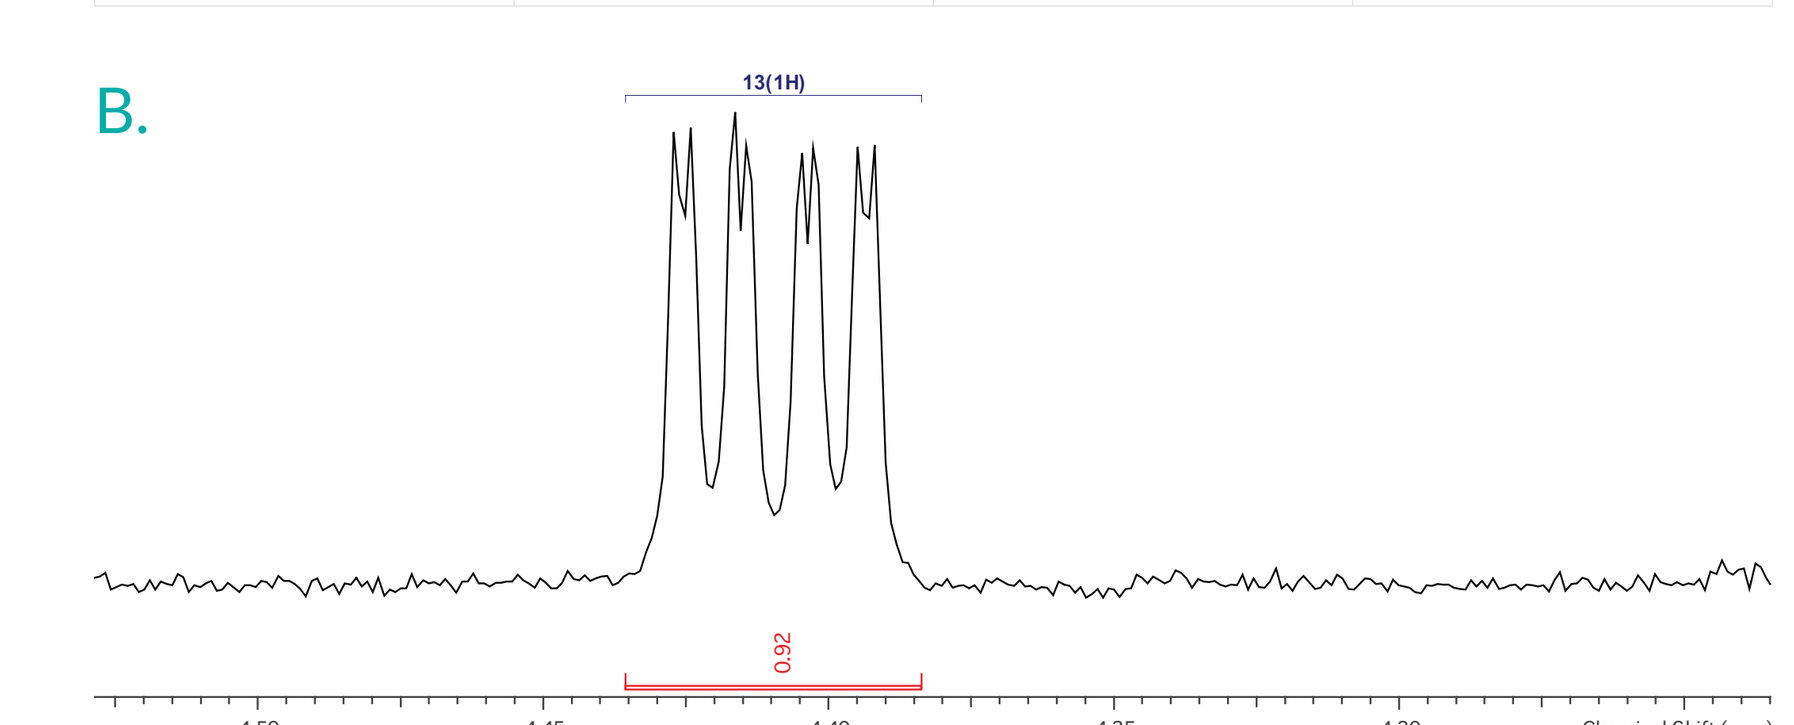
13(1H)
Chemical Shift (ppm)

Figure 5. A. J-coupling of all epoxide protons was in the range of 2.1-2.3 Hz. This suggests that both epoxides are in the *trans* configuration.

Figure 5. B. J-coupling of 13-C**H**-O suggests *trans* configuration relative to the 11,12-epoxide.

Conclusions and Ongoing Work

As a result of our studies, the structure of a newly discovered arachidonic acid metabolite was established as 8,11-DiEp-13-HEDE.

The specific stereochemistry of the purified lipid could not be established from the current data nor could the specific stereochemistry of the platelet-derived product. The NMR data unambiguously establishes the structure of the purified lipid from the autoxidation reaction to be 8,11-DiEp-13-HEDE. The data also clearly demonstrates that the epoxides exist in the *trans* configuration and the relative stereochemistry of the 13-hydroxy and 11,12-epoxide is *trans*. The LC and MS comparison of the platelet-derived product and autoxidation product also suggests that the platelet-produced lipid is related to this structure rather than the previously reported dioxolane structure.

Additional efforts are ongoing to establish the remaining relative and absolute stereochemistry of the platelet-derived oxylipin. Due to the COX-1-based origins of the lipid in platelets, we presume that C-11 is a single enantiomer, but cannot assign any absolute configurations. Early biological testing was limited by the small amount of native 8,11-DiEp-13-HEDE available from platelet incubations, but this can now be interrogated more widely using oxidatively sourced material.